CHARACTERIZATION OF ADCS AND MABS BY DIRECT COUPLING OF CAPILLARY ISOELECTRIC FOCUSING WITH MASS SPECTROMETRY

Gianni Vandenborre[1]; Jasmin Schairer[1]; Esther Devloo[1]; Thu Ba Nguyen[2]; Tao Bo[3]; Tiemin Huang[3]; Mabelle Meersseman[1]; Pat Sandra[1]; Koen Sandra[1]

[1]RIC group, Kortrijk, Belgium;
[2]Isogen Life Science BV, Utrecht, Netherlands;
[3]Advanced Electrophoresis Solutions Ltd, Cambridge, Canada

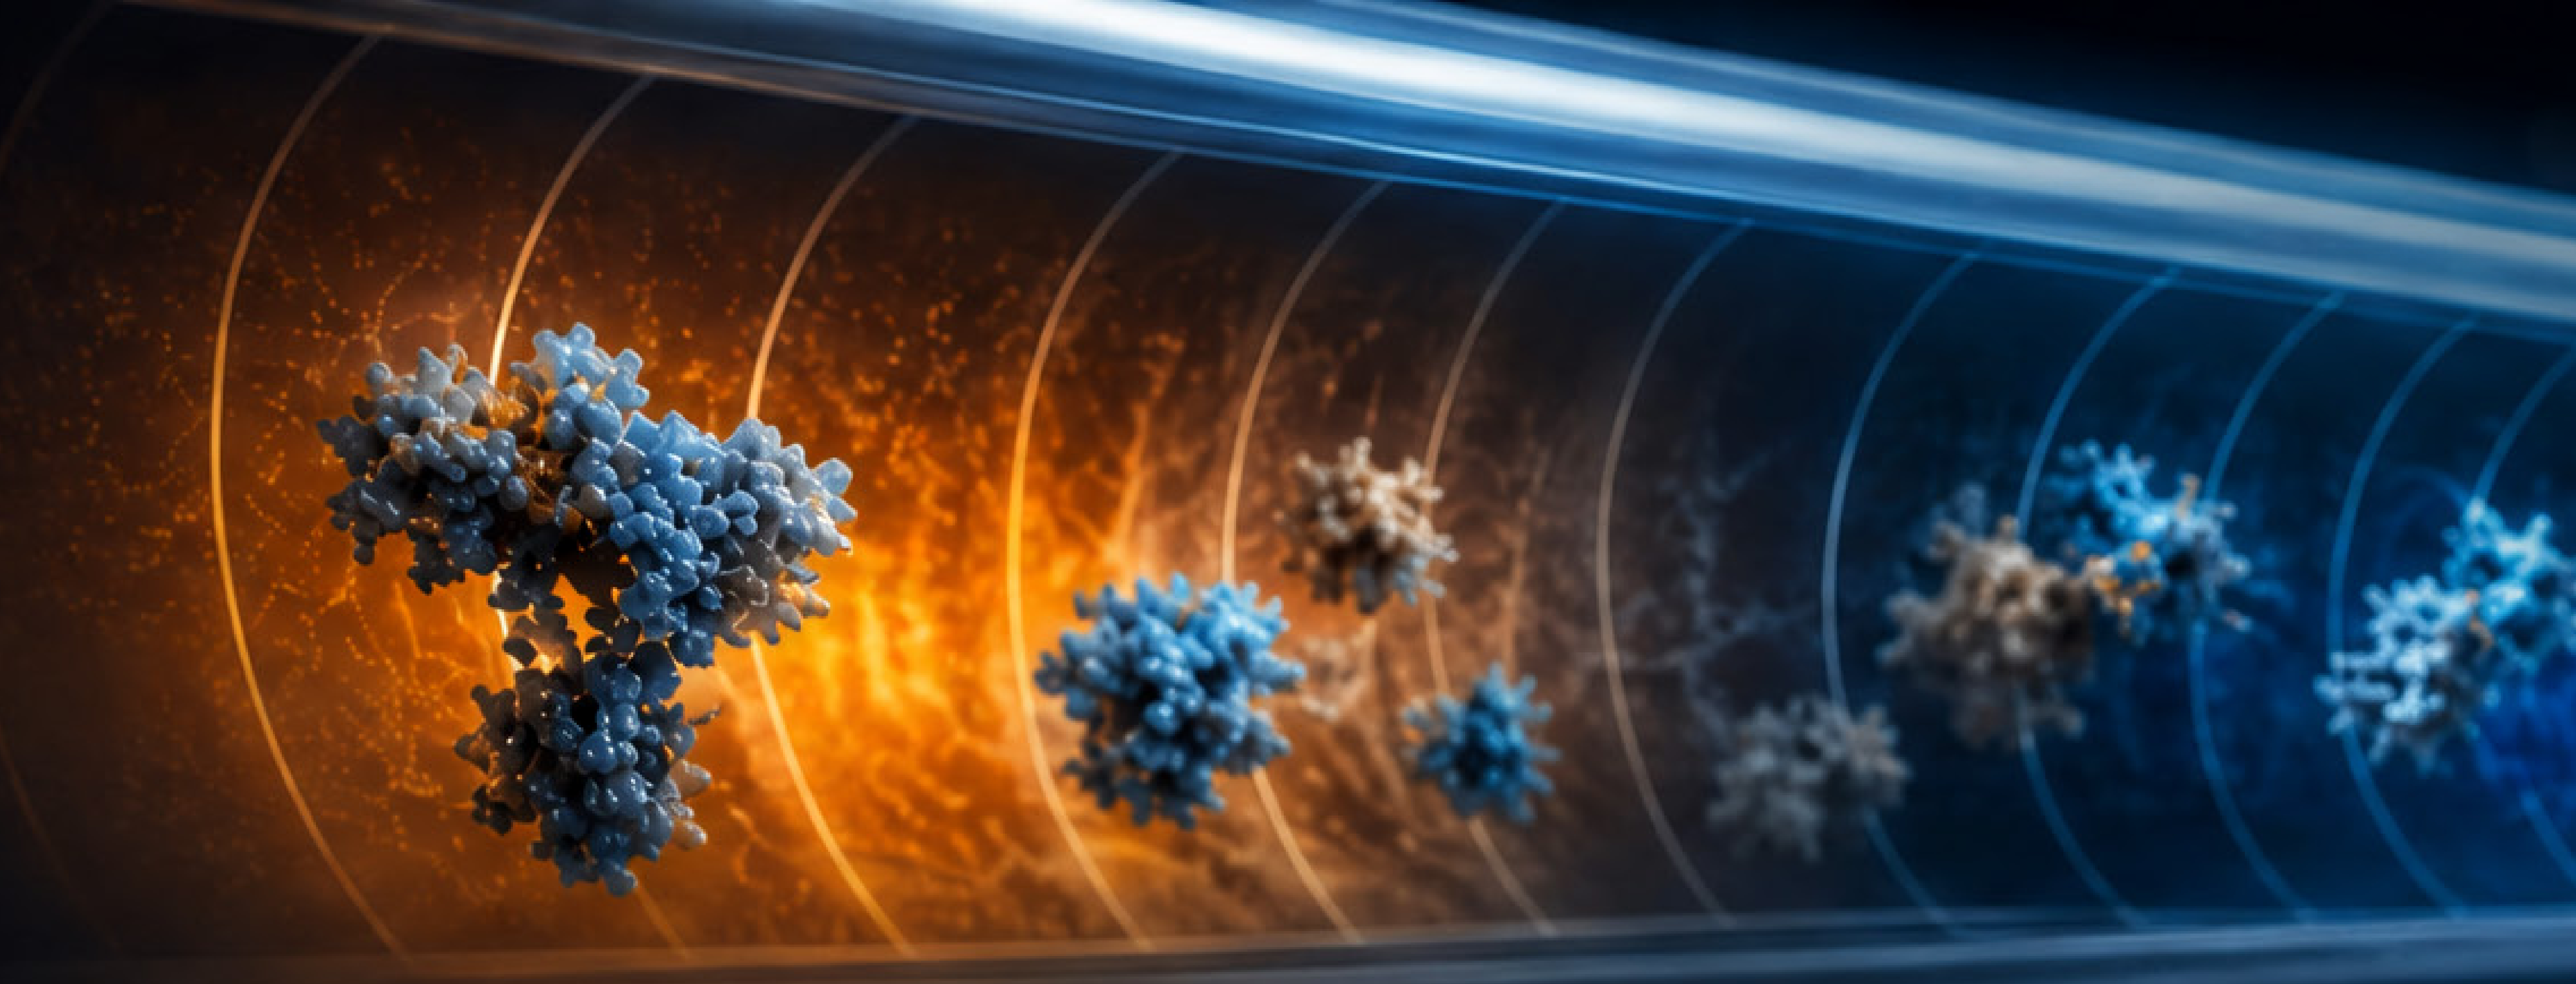

INTRODUCTION

Therapeutic proteins constitute a rapidly expanding class of biopharmaceuticals designed to treat a wide range of diseases, including cancer and autoimmune diseases.

A defining characteristic for all therapeutic proteins is their molecular heterogeneity caused by, among others, charge isoforms introduced during production and storage that may impact safety and efficacy. Capillary isoelectric focusing (cIEF) has become the method of choice when it comes to charge variant separation, however, direct coupling of cIEF to MS is challenged by the use of several MS incompatible components.

The introduction of MS-friendly alternatives and dedicated instrumentation has paved the way for the full exploration of cIEF-MS for the indepth characterization of therapeutic proteins such as antibody-drug conjugates (ADCs) and monoclonal antibodies (mAbs).

EXPERIMENTAL

Protein samples were concentrated to at least 40 mg/mL in water using 10K Amicon® ultracentrifugal filters. Then, samples were mixed with master mix, vortexed, and centrifuged before transfer to a test vial. A schematic overview for the direct coupling of icIEF with MS is shown in Figure 1.

MASTER MIX COMPOSITION	
Component	V (µL) / sample
Water	73
30% Formamide	100
AESlytes® SH 7-9	8
Arginine (500 mM)	2
Iminodiacetic acid (200 mM)	5
pI marker 4.65	1
pI marker 9.22	1

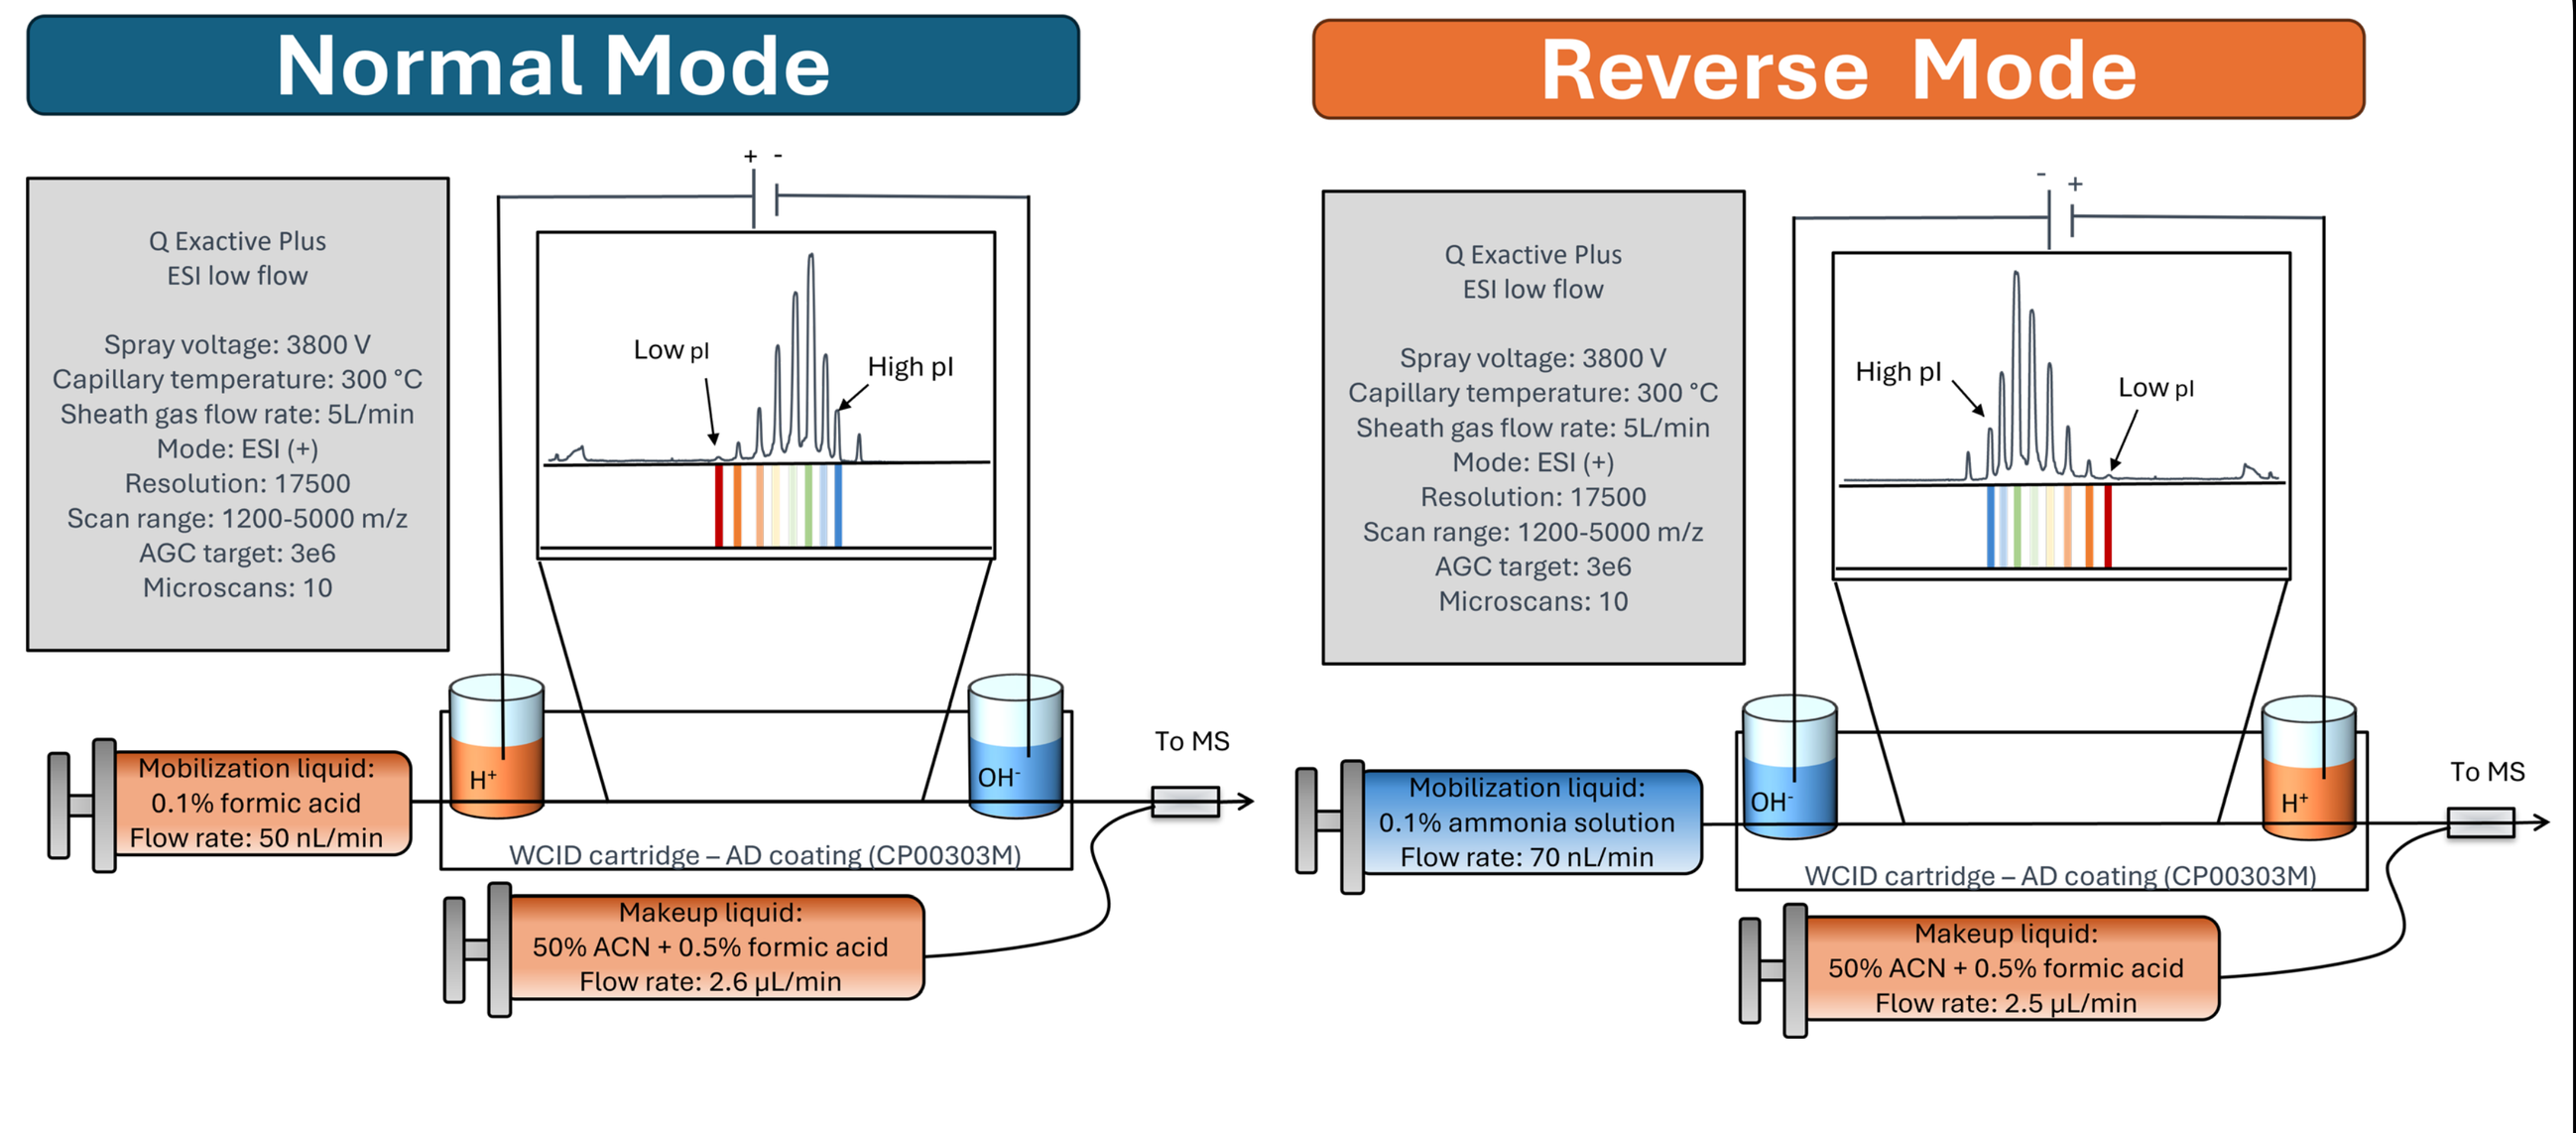
Normal Mode
Reverse Mode
Q Exactive Plus
ESI low flow
Spray voltage: 3800 V
Capillary temperature: 300 °C
Sheath gas flow rate: 5L/min
Mode: ESI (+)
Resolution: 17500
Scan range: 1200-5000 m/z
AGC target: 3e6
Microscans: 10
Low pI
High pI
Mobilization liquid: 0.1% formic acid Flow rate: 50 nL/min
Mobilization liquid: 0.1% ammonia solution Flow rate: 70 nL/min
WCID cartridge – AD coating (CP00303M)
Makeup liquid: 50% ACN + 0.5% formic acid Flow rate: 2.6 µL/min
Makeup liquid: 50% ACN + 0.5% formic acid Flow rate: 2.5 µL/min
To MS

Figure 1. Schematically setup of the direct icIEF-MS coupling.

Capillary coating
Capillaries were coated with an acrylamide derivative (AD) instead of the traditional fluorocarbon-based (FC) coating. This coating enables efficient focusing without the need for methylcellulose, which is incompatible with MS. In addition, capillaries with an internal diameter of 200 µm instead of 100 µm were used, allowing higher protein loads compared to conventional analytical icIEF. This increased load is required because the focused isoforms are diluted (>20x) by the make-up solution during mobilization towards MS (necessary for efficient electrospray ionization).

AESlytes®
AESlytes® are carrier ampholytes designed to limit MS background interference. Super high resolution (SH) AESlytes® were used (narrow range, pH 7 to 9).

Formamide
To maintain protein solubility at their respective pI, formamide was used instead of urea which is known to strongly suppress MS signals.

Polarity mode
Next to the normal polarity mode, the CEInfinite system enables a reversed polarity icIEF configuration (Figure 1). This mode is of interest because it prevents cross-contamination of acidic isoforms during mobilization towards MS by more dominant isoforms.

Download poster.
DRAFT VERSION - DO NOT PRINT!

RESULTS & CONCLUSIONS

Kadcyla® (trastuzumab emtansine, or T-DM1) is a first-in-class ADC designed for the treatment of HER2-positive breast cancer. It combines the targeted precision of trastuzumab with the cytotoxicity of DM1. Trastuzumab and DM1 are combined by a non-cleavable MCC linker, which ensures the toxic payload remains attached and only becomes released in the cancer cell once internalized.

The distinct isoforms observed in the cIEF profile of Kadcyla® were clearly correlated with species exhibiting increasing drug-to-antibody ratios (DARs). This is expected, as the MCC linker forms a stable covalent bond with the primary amino group of lysine, leading to progressive changes in pI with increasing levels of conjugation. This relationship was consistently confirmed using both normal and reversed polarity (Figures 2 and 3). In addition to the presence of the many DAR species, an additional mass variant was found linked to a population of Kadcyla® containing the unconjugated MCC linker without DM1 payload. This is the result of an incomplete conjugation process during manufacturing which is a two-step process. First trastuzumab is coupled to MCC via an NHS ester to lysine (step 1), and afterwards this intermediate reacts with cytotoxin DM1 (payload coupling, step 2).

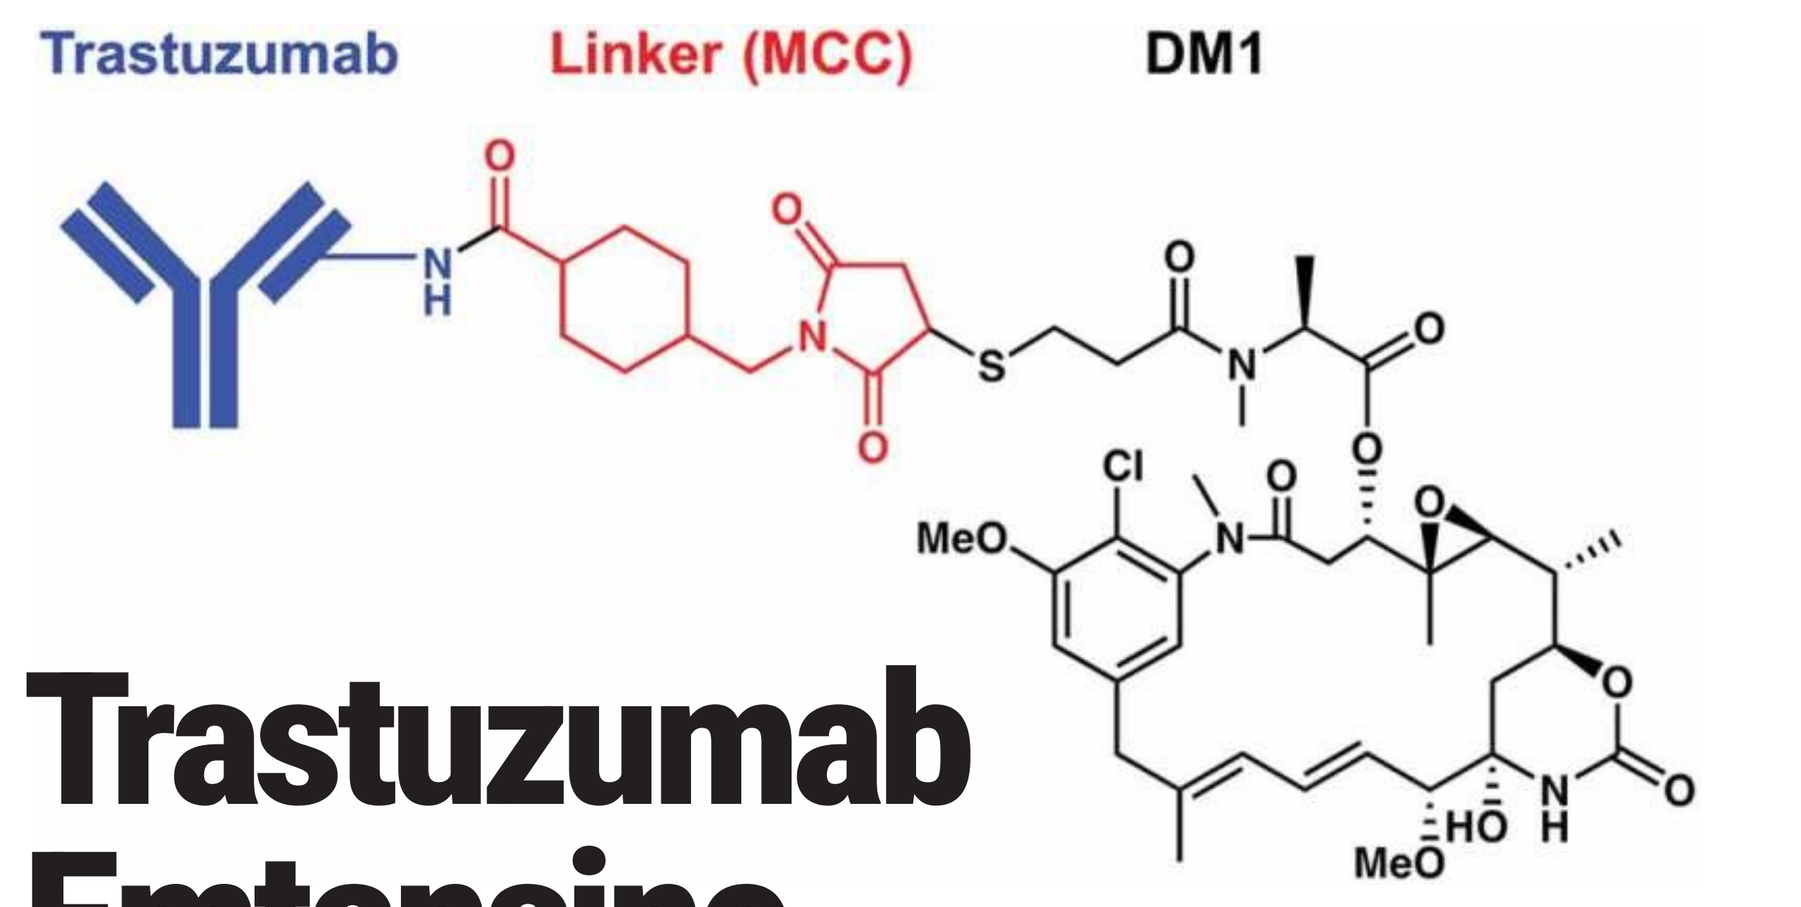
Trastuzumab
Linker (MCC)
DM1

Trastuzumab Emtansine

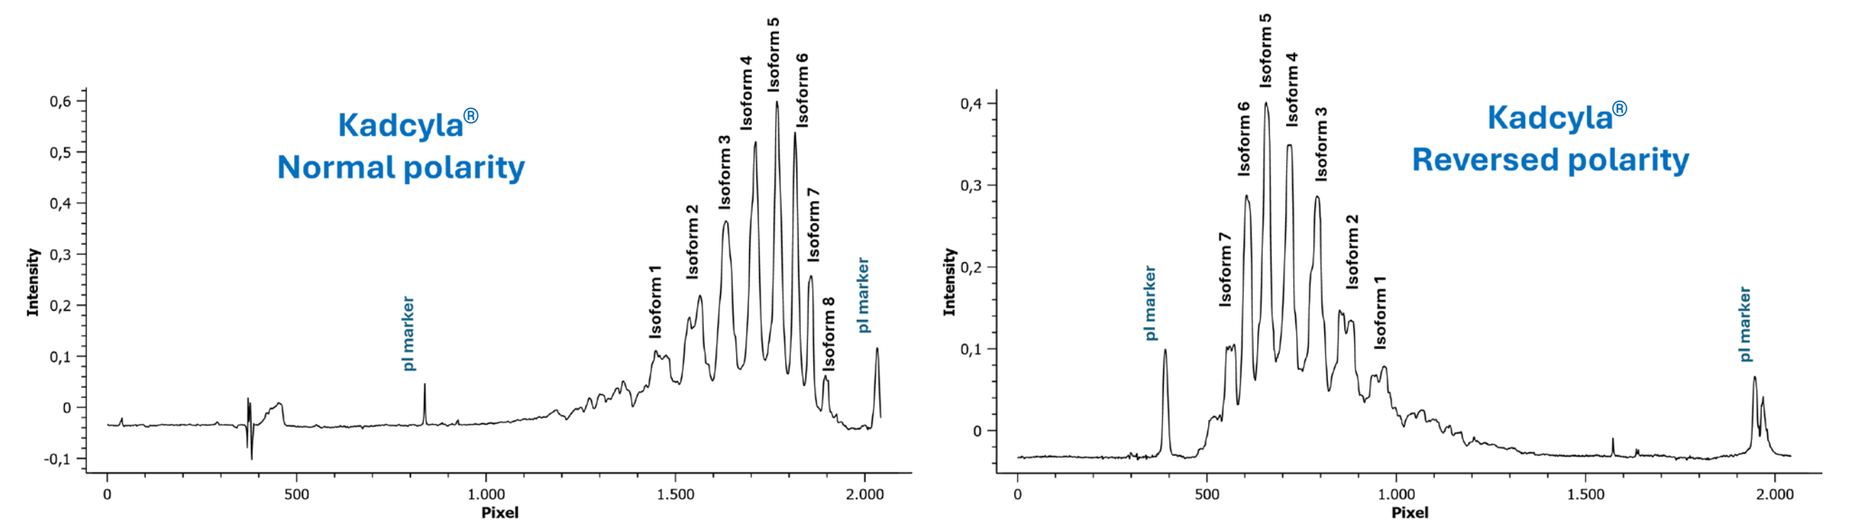
Kadcyla®
Normal polarity
Kadcyla®
Reversed polarity
Pixel
Intensity
pI marker

Figure 2. cIEF UV (280 nm) profiles of Kadcyla® using normal (A) and reversed polarity (B).

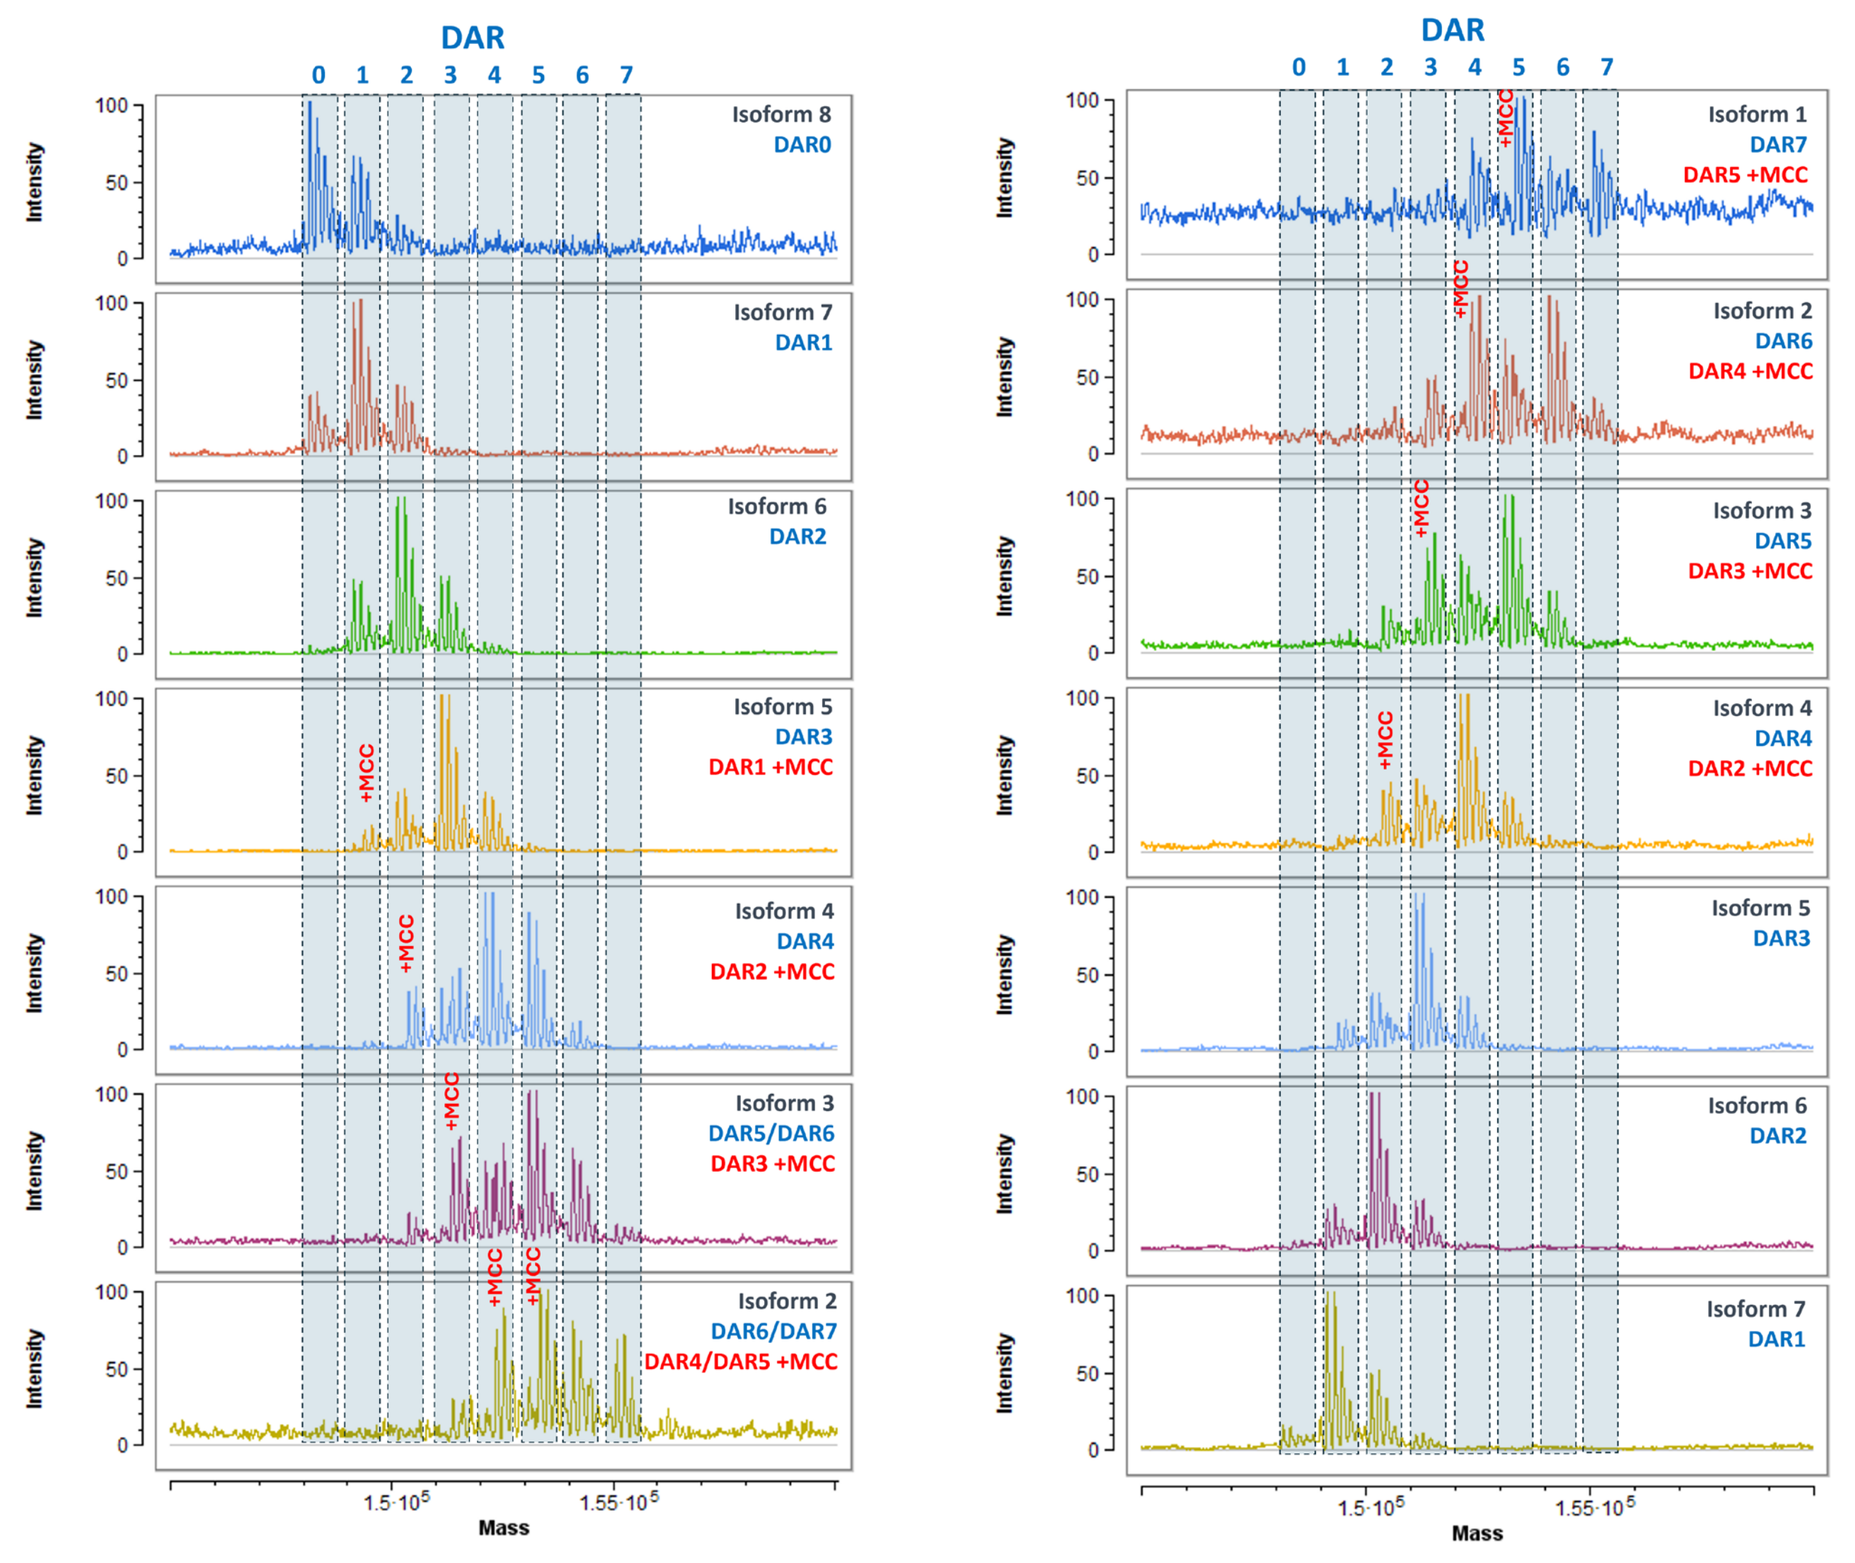
DAR
0 1 2 3 4 5 6 7
Isoform 8 DAR0
Isoform 7 DAR1
Isoform 6 DAR2
Isoform 5 DAR3 DAR1 +MCC
Isoform 4 DAR4 DAR2 +MCC
Isoform 3 DAR5/DAR6 DAR3 +MCC
Isoform 2 DAR6/DAR7 DAR4/DAR5 +MCC
Isoform 1 DAR7 DAR5 +MCC
Isoform 2 DAR6 DAR4 +MCC
Isoform 3 DAR5 DAR3 +MCC
Isoform 4 DAR4 DAR2 +MCC
Isoform 5 DAR3
Isoform 6 DAR2
Isoform 7 DAR1
Intensity
Mass

Figure 3. Deconvoluted MS spectra for the different isoforms present in the cIEF profile of Kadcyla® obtained by normal (left) and reversed polarity (right). Some isoforms contained two populations of Kadcyla®,: the DAR variant (with corresponding glycoforms) and a variant containing a modified MCC linker.

Cetuximab (brand name Erbitux®) is a recombinant human/mouse chimeric mAb that targets the epidermal growth factor receptor (EGFR), thereby inhibiting proliferation of cancers cells. Cetuximab exhibits glycosylation in both its Fc and Fab regions giving rise to a plethora of glycoforms and an exceptional charge heterogeneity. Lupitux® is a biosimilar for Erbitux® produced in a different host.

Although Lupitux® is developed as a biosimilar for Erbitux® clear differences were found in both basic and acidic variants (Figure 4). The abundance of C-terminal lysine is much higher for Erbitux® compared to Lupitux® (isoform 1 and 2, Figure 5). In addition, differentiation was found in the acidic mass variants (isoform 4-8, Figure 5). While Erbitux® is decorated with N-glycans containing (multiple) N-glycolyl neuraminic acid (Sg), the acidic species in the biosimilar contain (multiple) N-acetyl neuraminic acid (Sa) species. Moreover, Erbitux® is decorated with galactose-α-1,3-galactose (Ga) while this motif is absent in Lupitux®. This glycosylation difference can be attributed to the cell line used to recombinantly produce cetuximab, i.e. mouse myeloma cells (SP2/0) for Erbitux® and Chinese hamster ovary (CHO) cells for Lupitux®.

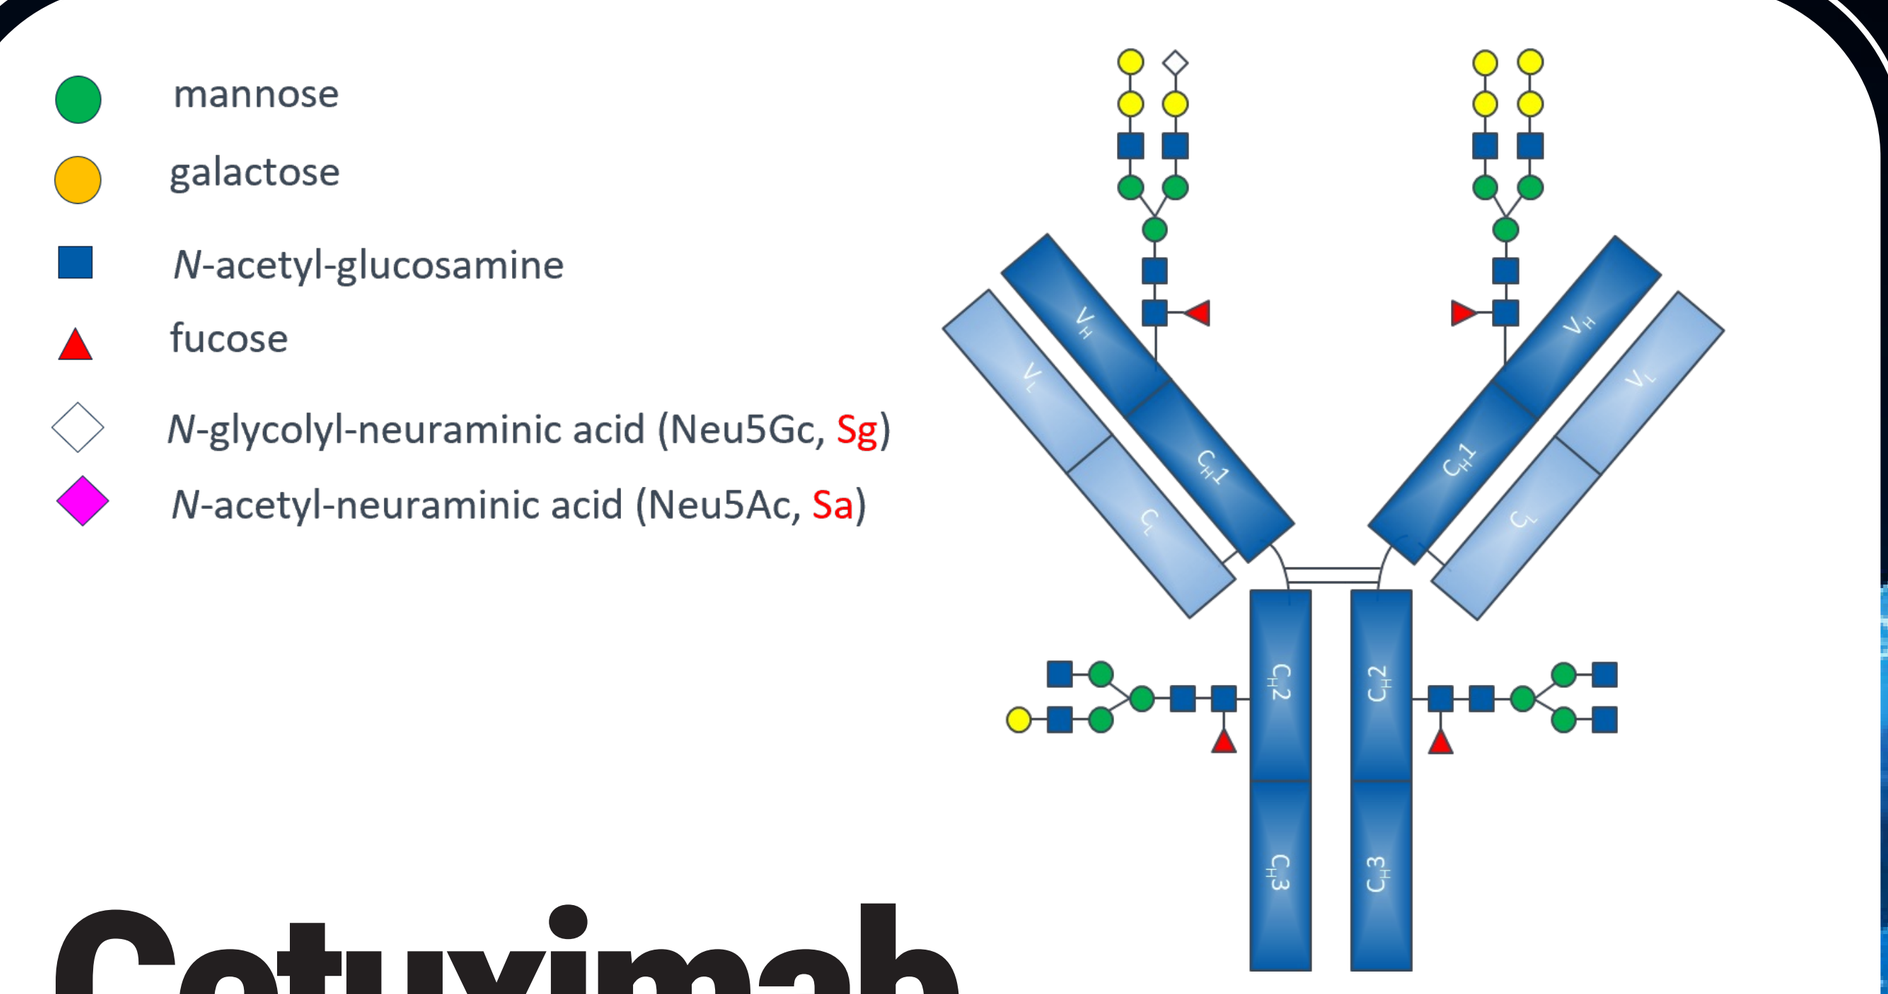
mannose
galactose
N-acetyl-glucosamine
fucose
N-glycolyl-neuraminic acid (Neu5Gc, Sg)
N-acetyl-neuraminic acid (Neu5Ac, Sa)

Cetuximab

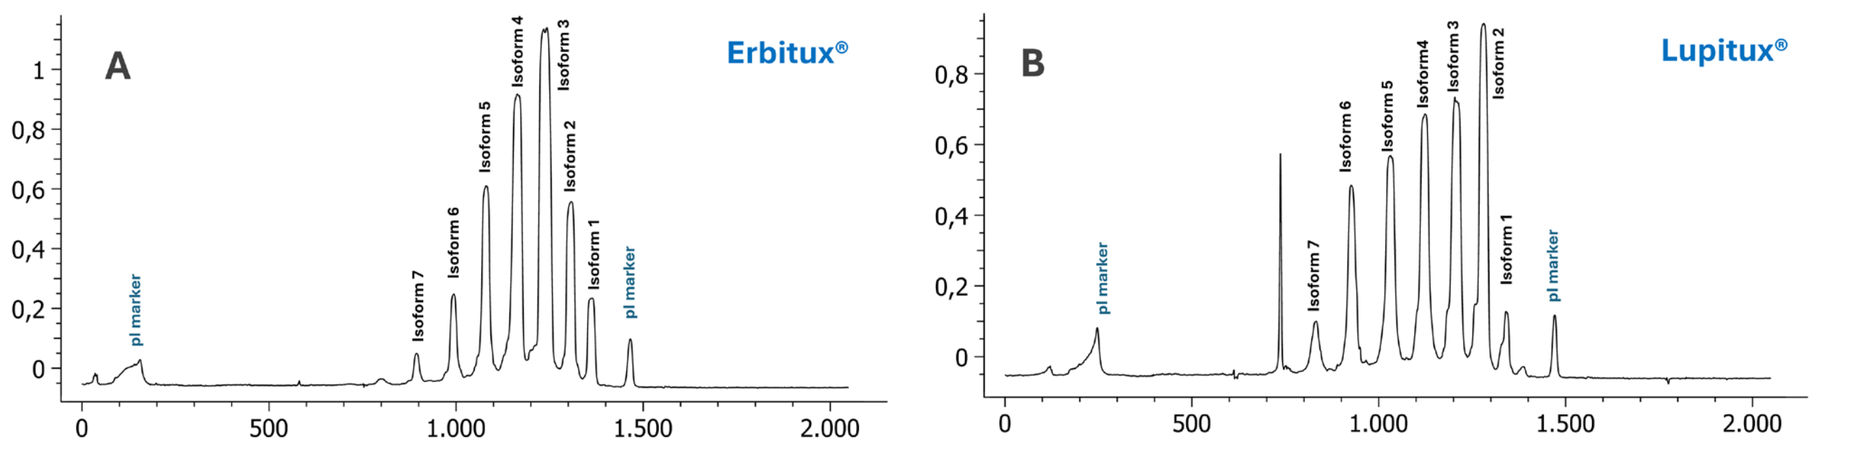
A
Erbitux®
B
Lupitux®
pI marker

Figure 4. cIEF UV (280 nm) profiles of Erbitux® (A) and Lupitux® (B) in reversed polarity.

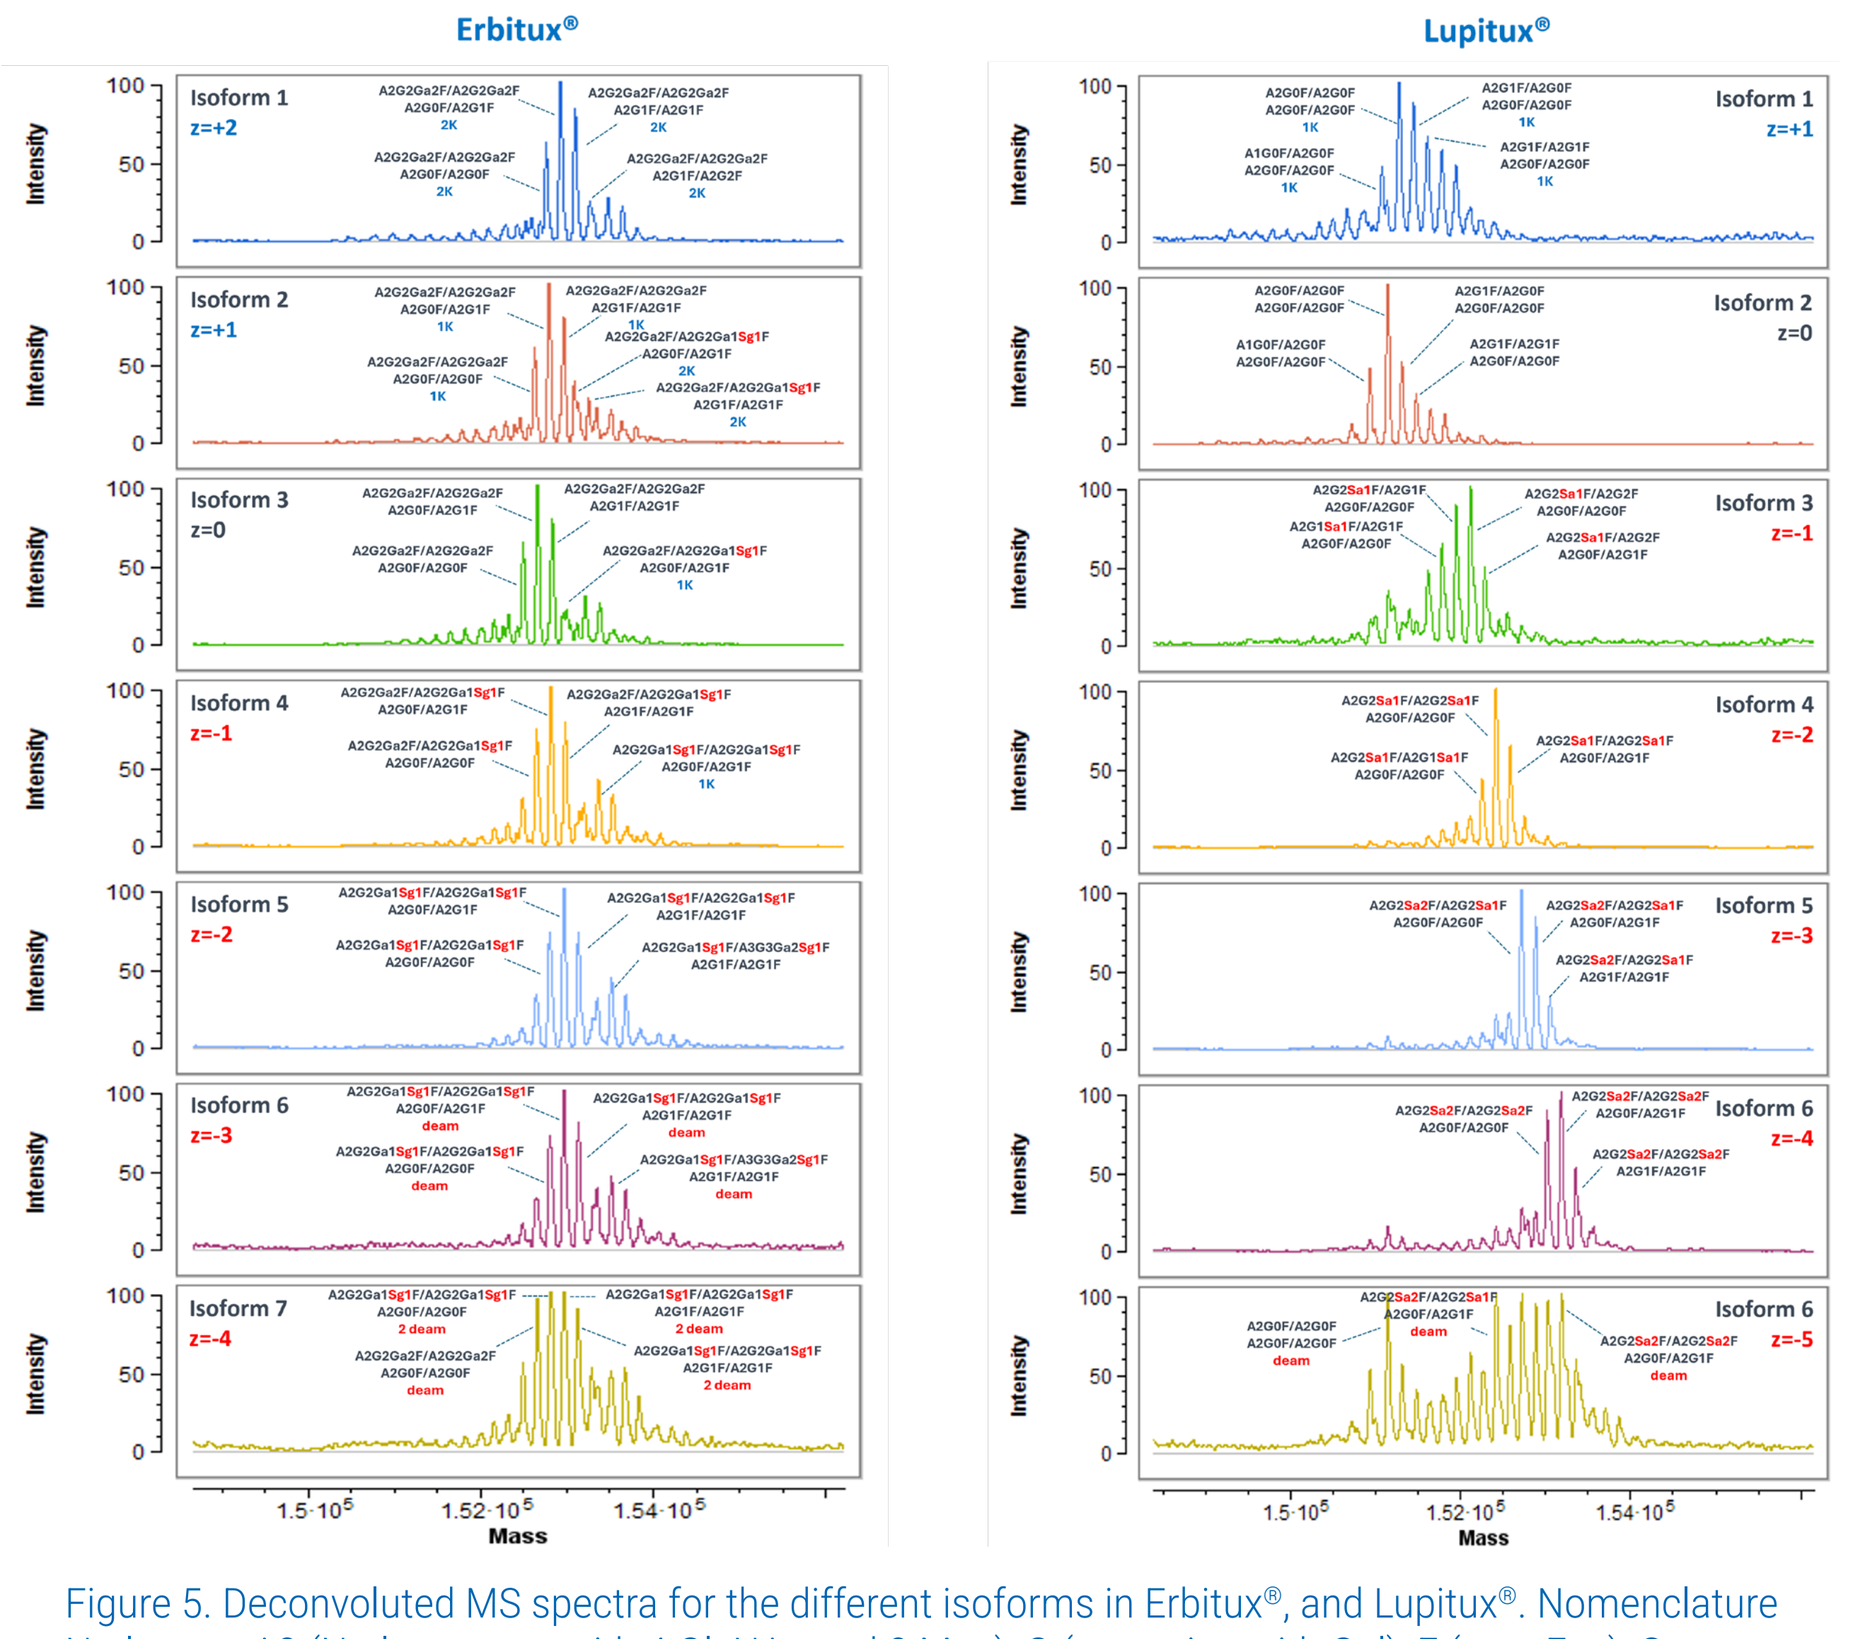
Erbitux®
Lupitux®
Intensity
Mass

Figure 5. Deconvoluted MS spectra for the different isoforms in Erbitux®, and Lupitux®. Nomenclature N-glycans: A2 (N-glycan core with 4 GlcNAc and 3 Man), G (extension with Gal), F (core Fuc), Ga (extension with second Gal), Sa (Neu5Ac), Sg (Neu5Gc) K (C-terminal lysine).